Cognitive enhancement and immune modulating properties of an antioxidant-rich microalgae extract, AFAninPlus™

Gitte S. Jensen[1], Steve G. Carter[1], Kimberlee A. Redman[1], Kathleen F. Benson[1], Axel Ehmann[2], Jesse Guthrie[2], Jim Turner[2], Christian Drapeau[2].
[1]NIS Labs, 1437 Esplanade Avenue, Klamath Falls, Oregon.
[2]Desert Lake Technologies LLC, 3735 Washburn Way, Klamath Falls, Oregon.

Objective

The purpose of this study was to evaluate the antioxidant, immune modulating, and cognitive effects of AFAninPlus™.

AFAninPlus™

AFAninPlus™ is an extract from the wild-harvested certified organic microalgae *Aphanizomenon flos aquae* (AFA). This microalgae is the predominant species in the hypereutrophic Upper Klamath Lake, Oregon, and at certain times of the season grows as a monoculture that can be harvested for consumption (Figure 1). At other times during the growing season, other algal species contribute to some extent to the biomass, and during that time AFA can be harvested using a selective harvesting technology (Patent pending) [1].

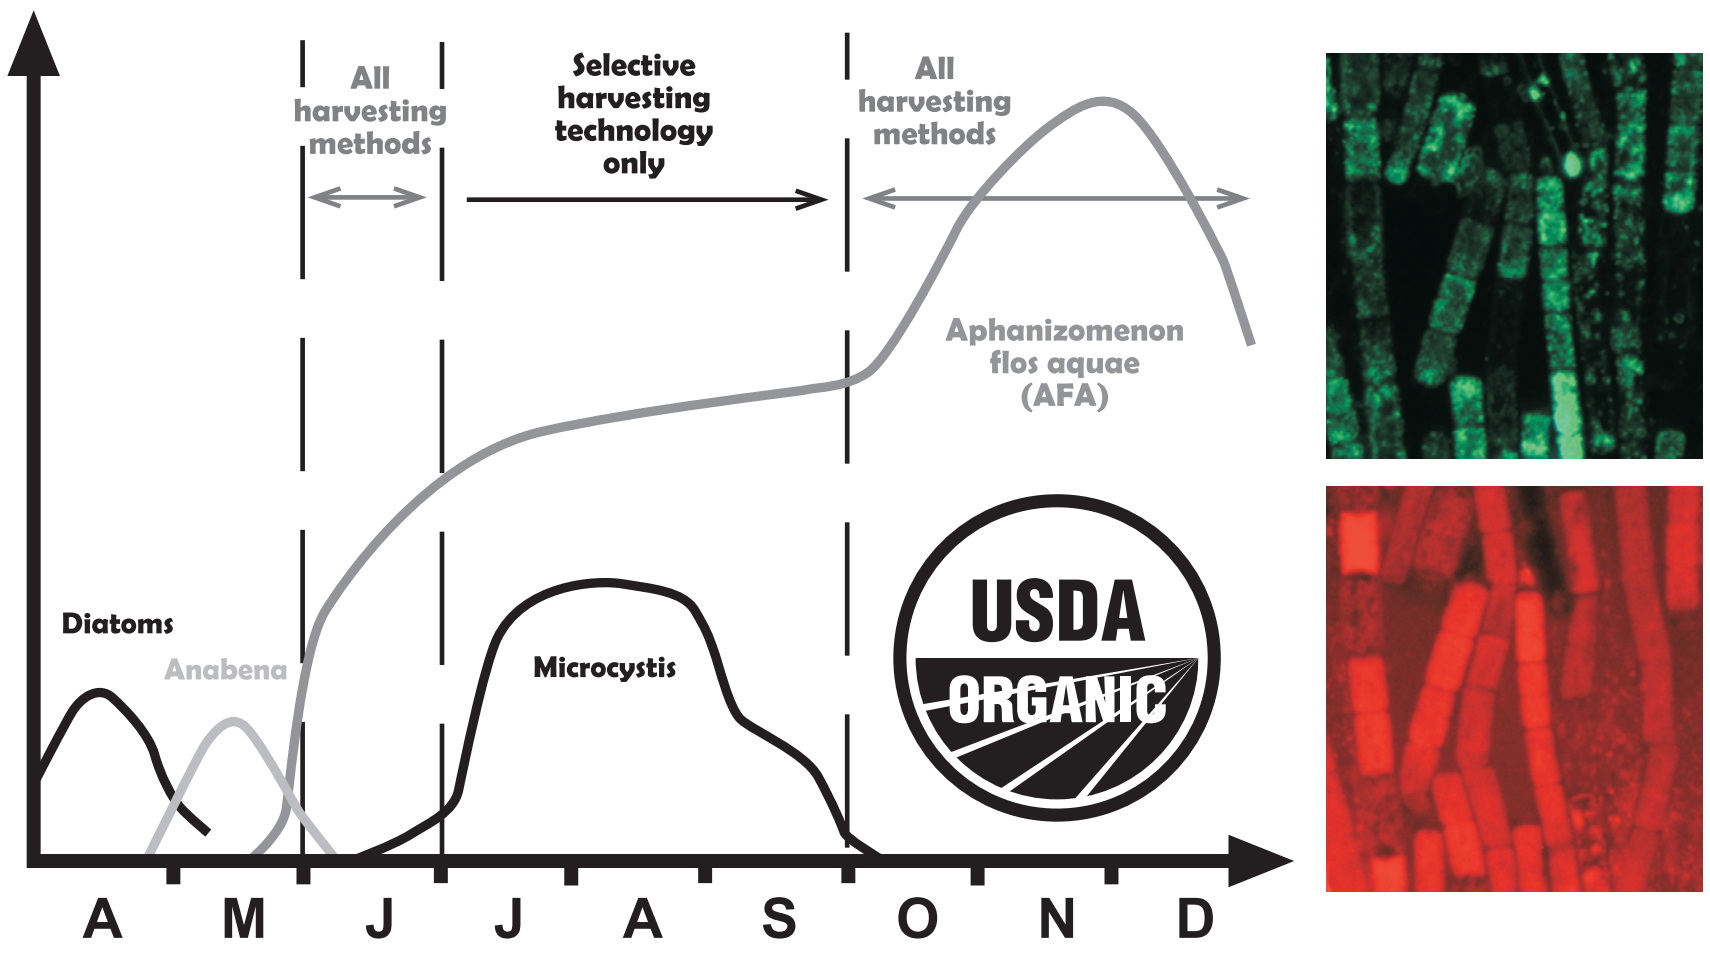
All harvesting methods
Selective harvesting technology only
All harvesting methods
Aphanizomenon flos aquae (AFA)
Diatoms
Anabena
Microcystis
USDA
ORGANIC
A
M
J
J
A
S
O
N
D

Figure 1. *Variations in the predominant plankton species in Upper Klamath Lake across the seasons from April to December. The top right micrograph shows AFA cells using light microscopy, and the bottom right micrograph shows AFA cells fluorescing in red. The red fluorescence is caused by the content of the antioxidant and anti-inflammatory compound Phycocyanin (PC).*

The AFAninPlus™ extract is enriched from the crude AFA biomass to allow a more concentrated content of specific compounds. Those compounds responsible for the effects on cognition and immune support are phenylethylamine, Phycocyanin, and complex polysaccharides.

Phenylethylamine (PEA) content is unique to AFA among edible microalgae and is also produced by the human brain. PEA is an endogenous modulator of dopaminergic transmission and has been shown to be deficient in the nervous system of depressive individuals. In addition, PEA metabolites were reduced in the urine of depressed individuals [2-3]. Thus, daily consumption of PEA may alleviate the symptoms of depression [4]. PEA was also found to provide improvements in attention disorders [5] and has been shown to increase concentration and elevate mood. PEA is not normally present in plant-based products, unless generated by microbial fermentation. We must assume that AFA obtained the property of being able to synthesize PEA after its evolutionary divergence from other similar microalgae.

Phycocyanin (PC) is part of the light harvesting system in cyanophyta. PC is a known antioxidant and a selective COX-2 inhibitor [6], with cardio- and neuro-protective effects.

Complex polysaccharides present in AFAninPlus™ contribute to activation of immune cells, including Natural Killer (NK) cells [7].

Cognitive function

Cognitive function, including mental acuity, ability to focus, and mood, is negatively affected by inflammatory conditions and oxidative stress.

- Strong link between antioxidant levels and protection from neurodegenerative illnesses such as age-related dementia [8-11];
- Low Natural Killer (NK) cell cytotoxic activity, imbalances in cytokine profile, and low antioxidant status reported in people suffering from major depression and suicidal behavior [12-14];
- Direct anti-depressive effects of various antioxidant types [15-16];
- A recent article proposed that anti-inflammatory treatment strategies, used for treatment of immune dysfunction, may be highly useful in psychiatric treatment [17].

Therefore, a nutritional approach to a healthy cognitive function should include a combination of antioxidants, anti-inflammatory and neuro-protective substances.

In vitro testing

The in vitro testing confirmed that AFAninPlus™ inhibited the enzymatic activity of cyclooxygenase-2 (COX-2). Furthermore, antioxidants in AFAninPlus™ were able to cross the lipid bilayer cell membrane and protect living cells from intracellular oxidative stress. AFAninPlus™ was shown to activate NK cell expression of CD69, a marker that correlates with increased NK cell cytotoxicity, i.e. increased activity in terms of scavenging and killing transformed cells.

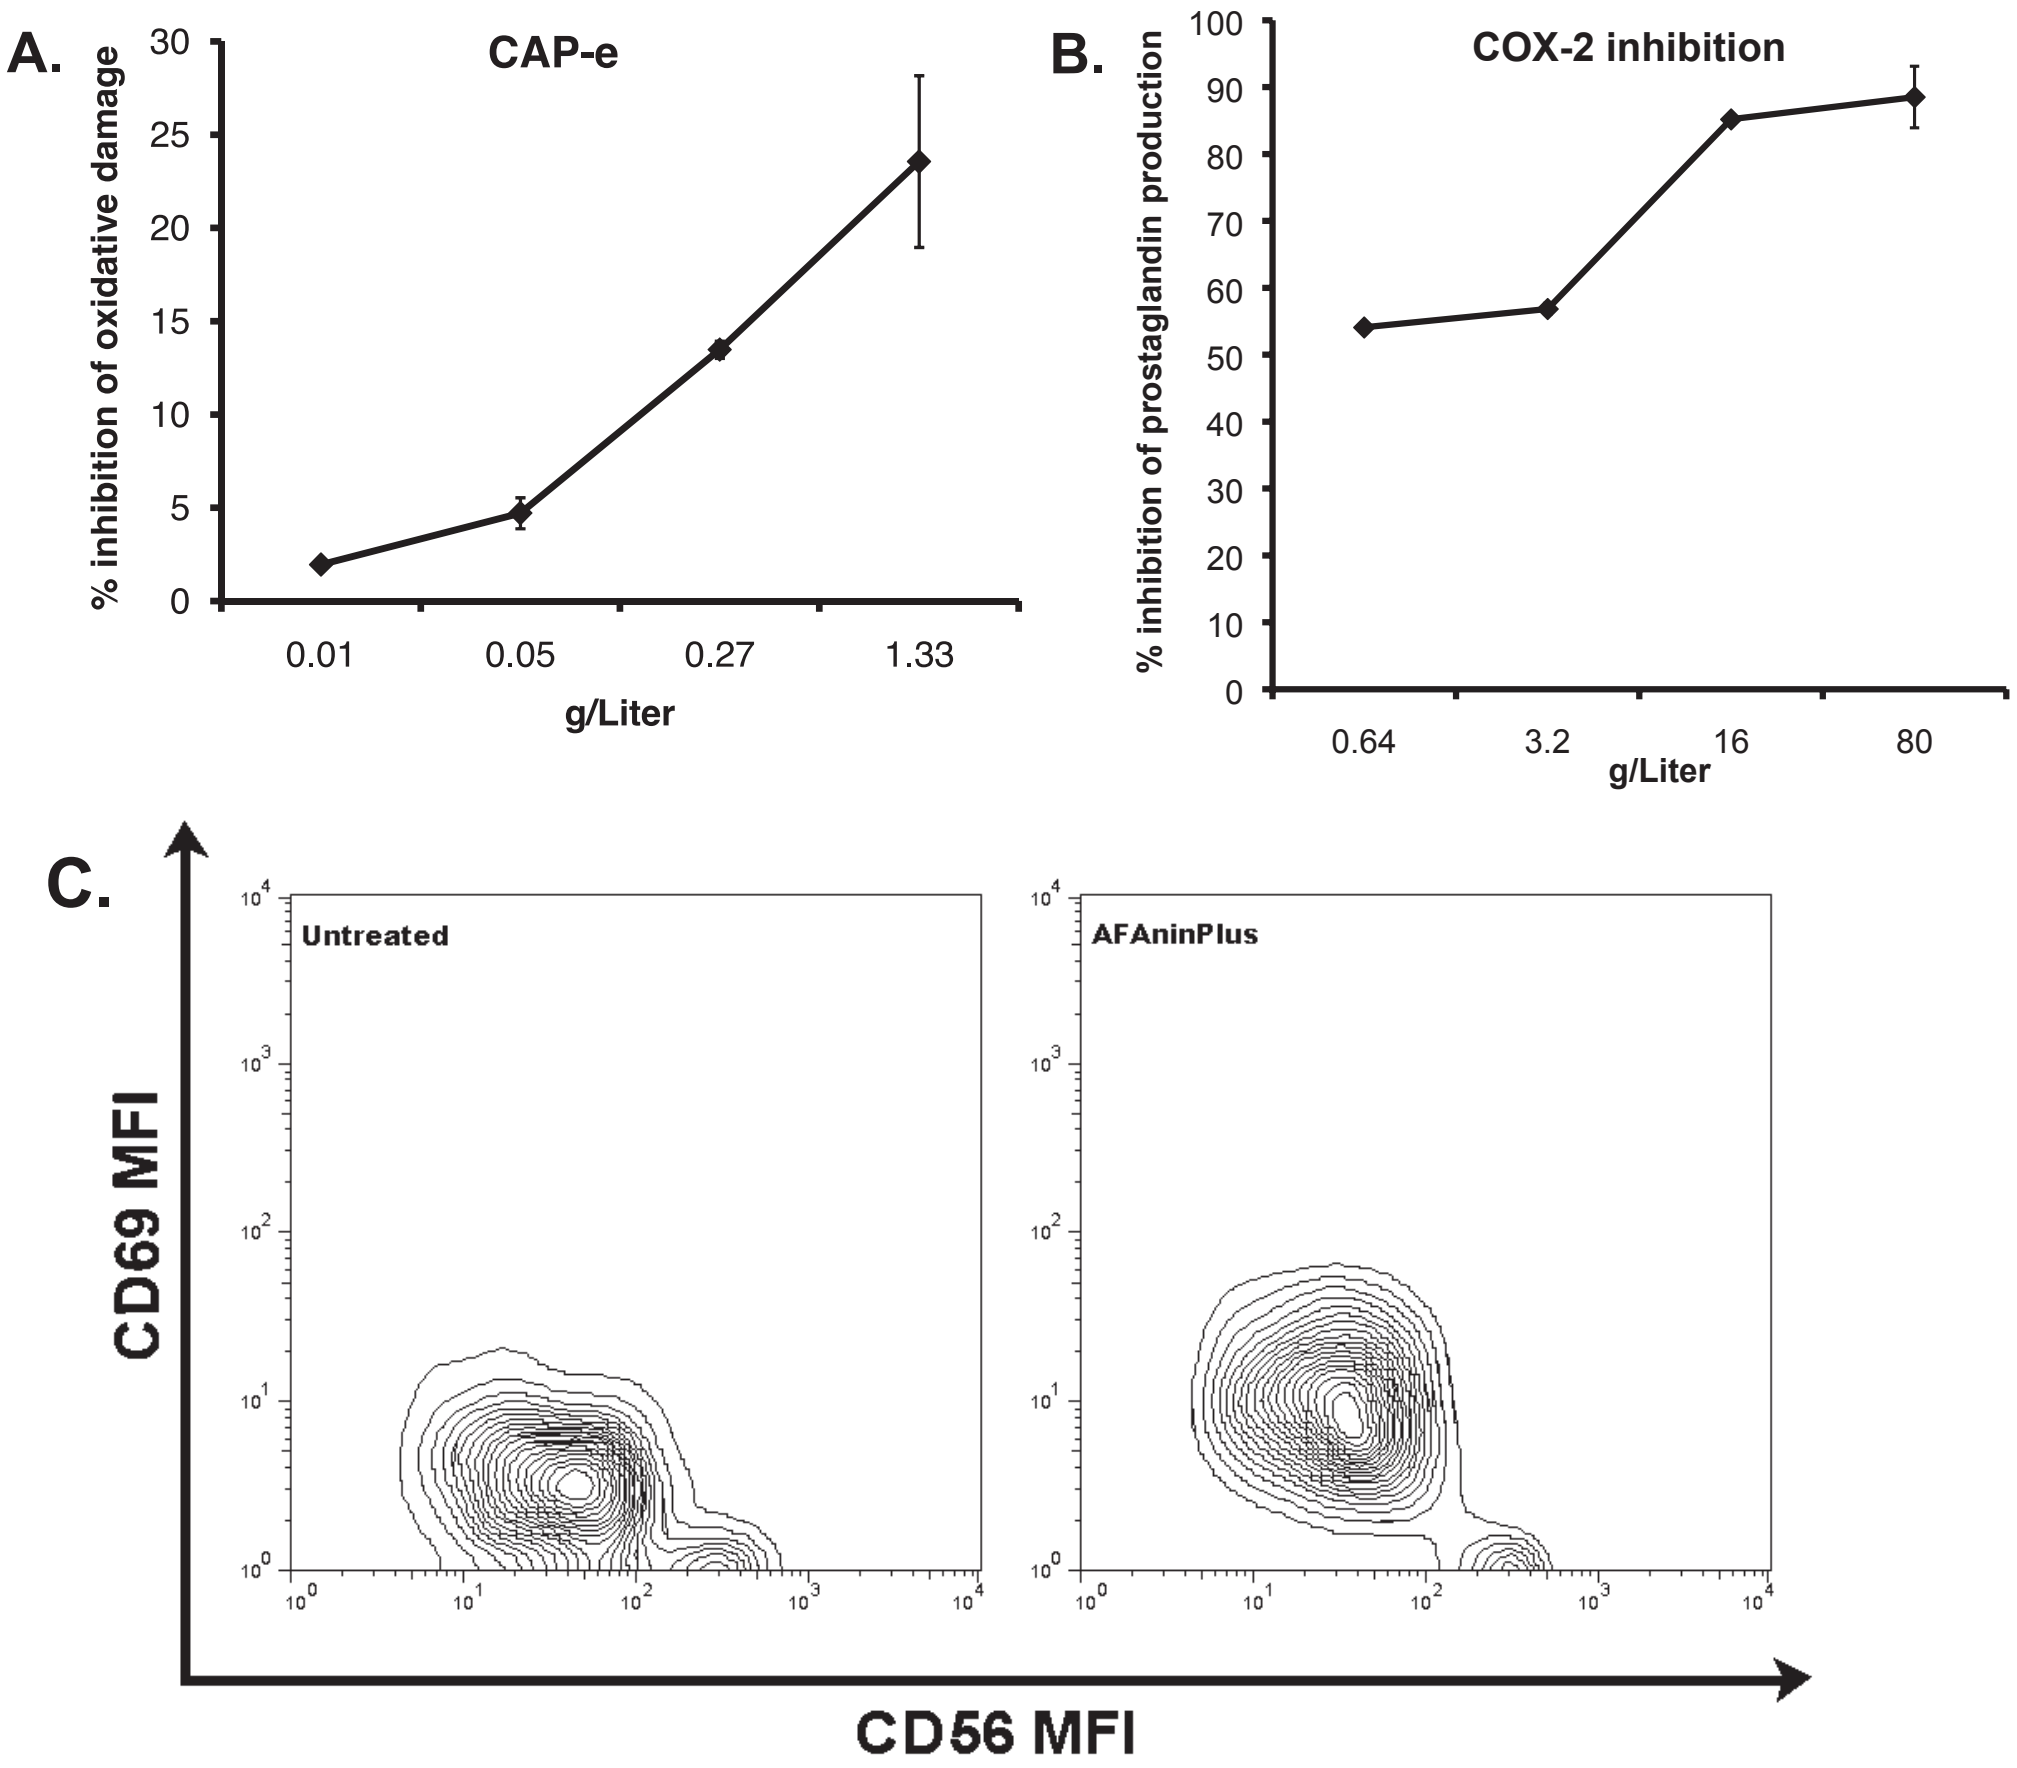
A.
CAP-e
% inhibition of oxidative damage
30
25
20
15
10
5
0
0.01
0.05
0.27
1.33
g/Liter
B.
COX-2 inhibition
% inhibition of prostaglandin production
100
90
80
70
60
50
40
30
20
10
0
0.64
3.2
16
80
g/Liter
C.
Untreated
AFAninPlus
CD69 MFI
CD56 MFI

Figure 2. *A) The antioxidant protection provided by AFAninPlus™ was evaluated by the CAP-e bioassay. B) Inhibition of COX-2 enzymatic activity was evaluated using a commercial microplate kit showing reduced production of prostaglandin. C) The capacity of AFAninPlus™ to activate natural killer (NK) cells in vitro was evaluated by culturing peripheral blood mononuclear cells (PBMC) in the absence versus presence of Interleukin-2, followed by immunostaining for CD3 negative, CD56 positive NK cells, and evaluating changes to the expression of the activation marker CD69. The expression of CD69 has been shown to correlate with the cytotoxic activity of NK cells, i.e. their active role in elimination of virus-infected and transformed cells.*

NIS LABS

DESERT LAKE TECHNOLOGIES

Clinical study on visual acuity and cognitive function

Three approaches were taken to evaluate effects of AFAninPlus™ on the central nervous system: 1) An interactive computer assessment of reaction time and ability to multitask, 2) Questionnaires on perceived ability to concentrate on the computer tasks, and 3) Evaluation of the physiological blind spot (PBS). The PBS is a traditional neurological test that indirectly measures the integrity of thalamo-cortical projections and therefore provides an objective measure of the ability of an individual to focus and concentrate. The eye has an anatomical blind spot where the nerve bundle and blood vessel enters the back of the eye, interrupting the retina. The physiological blind spot is the blank spot in our vision resulting from that interruption. The brain compensates for the PBS, and the size of the PBS is inversely related to cortical activity and mental acuity. A PBS that is larger than the anatomical blind spot is indicative of reduced visual acuity and an impaired ability to focus.

Table 1. Products* tested for effects on cognitive function and visual acuity.

Placebo	PEA-depleted AFA	2 gr	carrier: 5% glycerin in organic apple juice
Test product	AFAninPlus™	2 gr	carrier: 5% glycerin in organic apple juice

**Both products had the same appearance, taste, smell, and viscosity.*

A double-blinded placebo-controlled randomized pilot study involving 20 healthy adults was performed, after informed consent. Each study participant came to the clinic on two occasions, with at least one week wash-out period between visits. At each visit, baseline monitoring was immediately followed by ingestion of either placebo or product. At 10, 30, and 60 minutes after consumption, questionnaires and PBS assessment was administered to monitor changes in perceived and actual cognitive function and visual acuity.

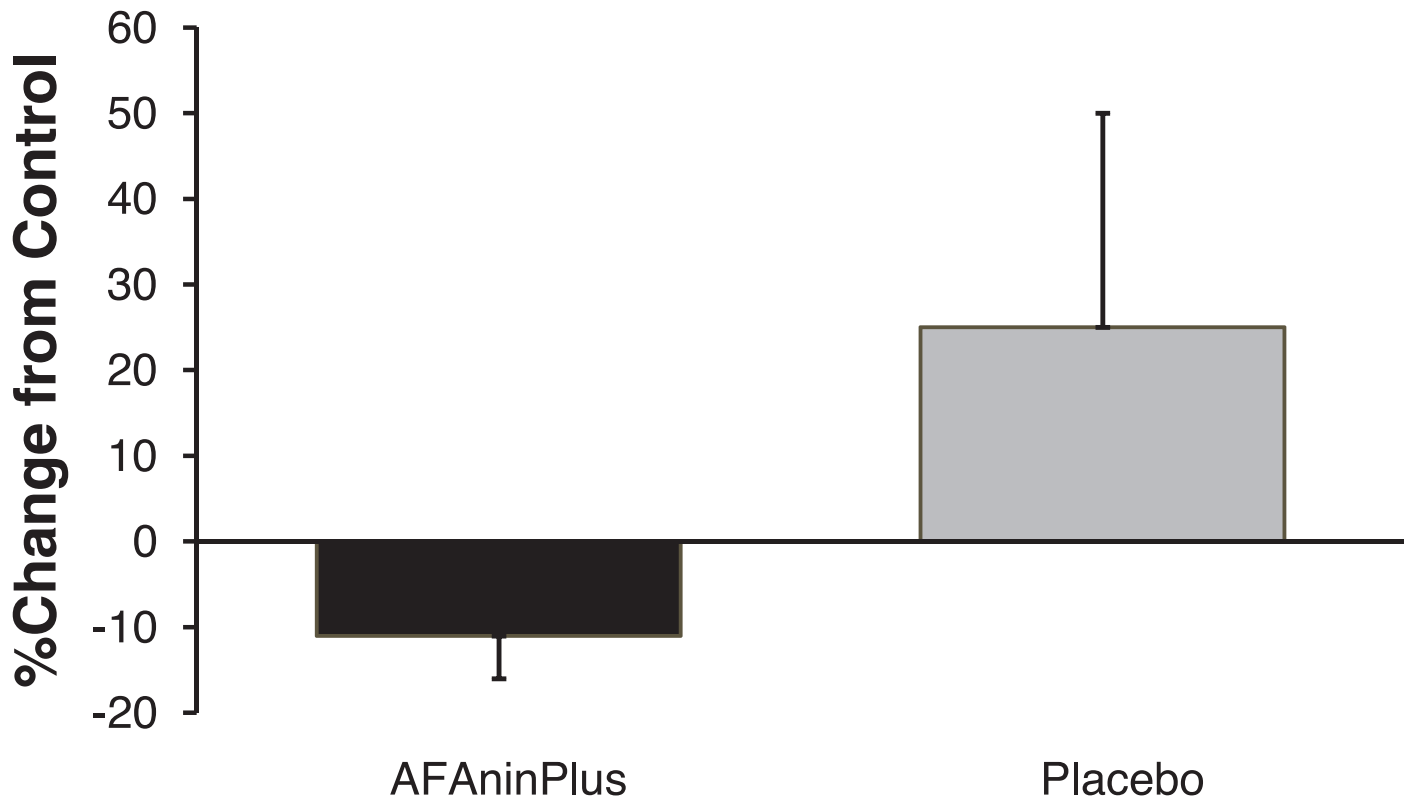

%Change from Control
60
50
40
30
20
10
0
-10
-20
AFAninPlus
Placebo

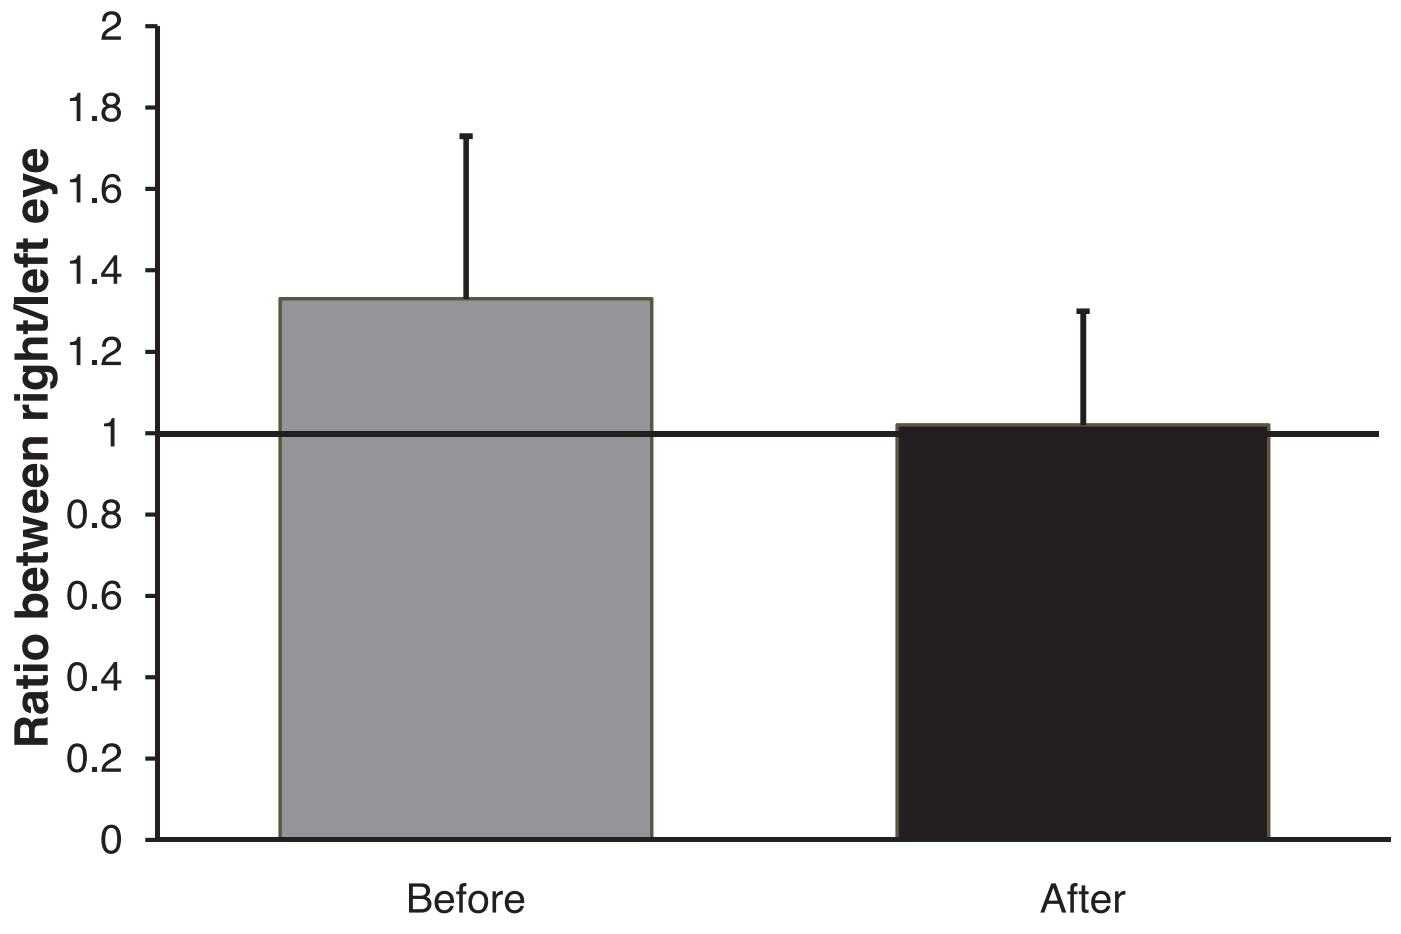

Ratio between right/left eye
2
1.8
1.6
1.4
1.2
1
0.8
0.6
0.4
0.2
0
Before
After

Figure 3. *A) The consumption of AFAninPlus™ resulted in rapid reduction of the PBS, indicating an improvement in visual acuity and cognitive function. Changes in the size of the PBS from consumption of either AFAninPlus™ or placebo were compared to the baseline value. The PBS for AFAninPlus™ was reduced about 11% (green bar), whereas the placebo PBS increased about 25% (tan bar). AfaninPlus™ (green bar) has a narrower standard deviation bar, indicating the response was similar across the study population. This is in contrast to the larger data spread in the placebo response, indicated by a larger error bar. The average improvement after consumption of AFAninPlus™ reached a high level of statistical significance (P<0.001).*
B) Consumption of AFAninPlus™ reduced the difference in the PBS between the right and left eye. A comparison of the right/left eye PBS was done by dividing the right PBS by the left PBS, so if they were equal the value would be “1”. At baseline, the PBS in the right eye was consistently larger than in the left eye, suggesting impaired inter-hemispheric coordination, this is of interest, as the right eye is dominant in over 70% of people.

In a separate study involving eight healthy adults, an interactive computerized method of assessment was employed. Selected sections of the Comprehensive Attention Battery (Neuropsych Works) and a touch-screen computer were used to evaluate rapid changes in reaction times, multitasking, and response to multi-sensory input (alternating auditory and visual commands to execute on touch screen). Questionnaires were used to evaluate each person’s subjective evaluation of focus and performance during the repeated tasks.
Consumption of AFAninPlus™ significantly improved reaction time and ability to multitask according to multi-sensory input, and also significantly improved the visual acuity by reducing the size of the PBS, when compared to placebo. The effect showed a bimodal pattern in most people, with a very rapid improvement seen within 2-10 minutes, and a more prolonged result, still effective at 1 hour after consumption.
Additional testing showed that at least part of the effect of AFAninPlus™ on cognitive function was due to rapid oral uptake of psychoactive compounds. When 2 grams AFAninPlus™ was held in the oral cavity for 2 minutes, and immediately expelled followed by an oral rinse, the effect on the visual acuity was of similar magnitude as when the product was consumed.

Conclusion

AFAninPlus™ has antioxidant, anti-inflammatory, and immune modulating properties in vitro, and has been shown to provide rapid improvement in cognitive function and visual acuity in vivo.

Acknowledgements

This study was conducted at NIS Labs, an independent contract research laboratory specializing in natural products testing. The study was sponsored by Desert Lake Technologies LLC.

References

1. Newman HW, Bowers J, Jones J: Apparatus for Extracting Material from Liquid and Methods therefore. US Provisional Patent Application No. 61/201,902
2. Sabelli HC, Fawcett J, Gusovsky F, Javaid JI, Wynn P, Edwards J, Jeffriess H, Kravitz H. Clinical studies on the phenylethylamine hypothesis of affective disorder: urine and blood phenylacetic acid and phenylalanine dietary supplements. (1986) J Clin Psychiatry 47(2):66-70.
3. Huebert ND, Schuurmans Schwach V, Richter G, Zreika M, Hinze C, Haegele KD. The measurement of beta-phenylethylamine in human plasma and rat brain. Anal Biochem (1994) 221(1): 42-7.
4. Sabelli HC, Fink P, Fawcett J, Tom C. Sustained antidepressant effect of PEA replacement. J Neuropsychiatry Clin Neurosci (1996) 8(2): 168-71.
5. Baker et al. (1991) Phenylethylaminergic mechanisms in attention deficit disorder. Biol Psychiatry 29(1):15-22.
6. Reddy CM, Bhat VB, Kiranmai G, Reddy MN, Reddanna P, Madyastha K. Selective inhibition of cyclooxygenase-2 by C-phycocyanin, a biliprotein from Spirulina platensis. Biochem Biophys Res Commun. (2000) 277(3):599-603.
7. Hart AN, Zaske LAM, Patterson KM, Drapeau C, Jensen GS: Natural Killer Cell Activation and Modulation of Chemokine Receptor Profile In Vitro by an Extract from the Cyanophyta *Aphanizomenon flos-aquae*. Journal of Medicinal Food (2007) 10(3): 435-441.
8. Willis LM, Shukitt-Hale B, Joseph JA. Recent advances in berry supplementation and age-related cognitive decline. Curr Opin Clin Nutr Metab Care. 2009 Jan;12(1):91-4.
9. Bastianetto S, Quirion R. Natural antioxidants and neurodegenerative diseases. Front Biosci. 2004 Sep 1;9:3447-52.
10. Mecocci P, Mariani E, Cornacchiola V, Polidori MC. Antioxidants for the treatment of mild cognitive impairment. Neurol Res. 2004 Jul;26(5):598-602.
11. Cumurcu BE, Ozyurt H, Etikan I, Demir S, Karlidag R. Total antioxidant capacity and total oxidant status in patients with major depression: impact of antidepressant treatment. Psychiatry Clin Neurosci. 2009 Oct;63(5):639-45. Epub 2009 Aug 10.
12. Doering LV, MartÌnez-Maza O, Vredevoe DL, Cowan MJ. Relation of depression, natural killer cell function, and infections after coronary artery bypass in women. Eur J Cardiovasc Nurs. 2008 Mar;7(1):52-8. Epub 2007 Aug 22.
13. Dhabhar FS, Burke HM, Epel ES, Mellon SH, Rosser R, Reus VI, Wolkowitz OM. Low serum IL-10 concentrations and loss of regulatory association between IL-6 and IL-10 in adults with major depression. J Psychiatr Res. 2009 Jul;43(11):962-9.
14. Kim YK, Lee SW, Kim SH, Shim SH, Han SW, Choi SH, Lee BH. Differences in cytokines between non-suicidal patients and suicidal patients in major depression. Prog Neuropsychopharmacol Biol Psychiatry. 2008 Feb 15;32(2):356-61.
15. Dimpfel W. Rat electropharmacograms of the flavonoids rutin and quercetin in comparison to those of moclobemide and clinically used reference drugs suggest antidepressive and/or neuroprotective action. Phytomedicine. 2009 Apr;16(4):287-94.
16. BinfarÈ RW, Rosa AO, Lobato KR, Santos AR, Rodrigues AL. Ascorbic acid administration produces an antidepressant-like effect: evidence for the involvement of monoaminergic neurotransmission. Prog Neuropsychopharmacol Biol Psychiatry. 2009 Apr 30;33(3):530-40. Epub 2009 Feb 11.
17. Berthold-Losleben M, Heitmann S, Himmerich H. Anti-inflammatory drugs in psychiatry. Inflamm Allergy Drug Targets. 2009 Sep;8(4):266-76.